C1-611

ACTIVITY OF PLAZOMICIN (PREVIOUSLY ACHN-490) AGAINST GRAM-POSITIVE AND GRAM-NEGATIVE BACTERIA IN THE PRESENCE OF PULMONARY SURFACTANT

LA FEENEY[1], R MATIAS[1], G MILLER[1], J BRUSS[2], E ARMSTRONG[1]

[1] Achaogen Inc., South San Francisco, CA USA [2] Medpace Inc., Cincinnati, OH USA

7000 Shoreline Court, Suite 371
South San Francisco, CA 94080
Phone: 650-266-1120, Fax: 650-266-1130
www.achaogen.com

ABSTRACT

Background: In order to treat bacterial pneumonia, antibiotics must maintain antimicrobial activity in the presence of pulmonary surfactant, a complex mixture of lipids and proteins. It has previously been reported that the activity of daptomycin, an antibiotic with Gram-positive activity, was inhibited in the presence of surfactant. Plazomicin (previously ACHN-490), a broad-spectrum aminoglycoside, is being developed to treat resistant bacterial pathogens. The *in vitro* activity of plazomicin and 6 comparator antibiotics was examined in the presence of varying levels of surfactant.

Methods: MICs were determined for plazomicin, amikacin, gentamicin, tobramycin, cefepime, and levofloxacin against 10 Gram-positive (*Staphylococcus aureus*) and 10 Gram-negative (5 each of *Pseudomonas aeruginosa* and *Klebsiella pneumoniae*) strains in the presence of 1%, 5%, and 10% bovine-derived lung surfactant, beractant (Abbott). Daptomycin and vancomycin were tested against *S. aureus* strains.

Results: The activity of plazomicin and comparator aminoglycosides against the 20 strains tested was not measurably affected by the addition of up to 10% surfactant (fold changes in MIC upon surfactant addition ranged from 0.25 to 4). The remaining antibiotics tested showed no consistent changes in potency with addition of surfactant causing 0.5- to 2-fold changes in MIC. In contrast, the activity of daptomycin against the *S. aureus* strains decreased from 16- to 32-fold in the presence of 1% surfactant and decreased at least 32-fold with 5% or 10% surfactant.

Conclusions: There was no surfactant-dependent decrease in plazomicin activity when either Gram-positive or Gram-negative bacteria were tested. With the exception of daptomycin, the activity of the comparator antibiotics was similarly unaffected by the presence of this surfactant.

Range of Fold Change in MIC with Surfactant

Antibiotic	Gram-Positive	Gram-Negative
Plazomicin	0.25 - 4	0.5 - 2
Daptomycin	16 - >64	N/A
Aminoglycosides (Amikacin, Gentamicin, Tobramycin)	0.25 - 4	0.25 - 4
Cefepime	0.5 - 2	0.5 - 2
Levofloxacin	0.5 - 2	1 - 2
Vancomycin	0.5 - 2	N/A

INTRODUCTION

Hospital-acquired pneumonia (HAP) is the second most frequent cause of hospital-acquired infections in the United States and the leading cause of morbidity and mortality from hospital-acquired infections. There is a need for new effective treatment options for HAP and other indications that has been exacerbated by the increasing prevalence of multi-drug resistant (MDR) pathogens.

Pneumonia is an infection of the bronchoalveolar spaces, which are normally bathed in alveolar lining fluid, a complex mixture of lipids and proteins, primarily surfactant, that serves to reduce surface tension in the alveolar regions. After failed clinical studies in pneumonia, daptomycin was shown to have diminished activity when tested in the presence of surfactant.[1] As a result of these findings, it is prudent for new antibiotics in development for the treatment of pneumonia to demonstrate the retention of antimicrobial activity in the presence of lung surfactant.

Plazomicin (previously ACHN-490) is a broad-spectrum aminoglycoside with differentiated activity against MDR Enterobacteriaceae, including those with *Klebsiella pneumoniae* carbapenemases (KPCs) and clinically relevant aminoglycoside modifying enzymes (AMEs).[2-4] Plazomicin is being developed as a first-line treatment for resistant bacterial pathogens and is currently in Phase II trials for complicated urinary tract infections. Additional clinical indications, including serious respiratory tract infections, are under investigation. The following study was done to help further confirm the utility of expanding the clinical profile of plazomicin to include serious respiratory tract infections (e.g., HAP and ventilator-associated pneumonia (VAP)). We evaluated the *in vitro* activity of plazomicin in the presence of increasing concentrations of pulmonary surfactant to demonstrate the retention of its antimicrobial activity. The panel of bacteria, both Gram-positive and Gram-negative, tested consisted of three species commonly responsible for nosocomial pneumonia infections (*Staphylococcus aureus*, *Pseudomonas aeruginosa* and *Klebsiella pneumoniae*). A total of 6 comparators representing 4 antibiotic classes were tested in parallel. These agents are frequently used either alone or in combination therapy to treat HAP and VAP. Additionally, the lipopeptide, daptomycin, was used as a positive control.

METHODS

Bacterial Strains: 10 Gram-positive strains including *S. aureus* ATCC 29213 and 9 clinical isolates. 10 Gram-negative strains composed of 5 *P. aeruginosa* (ATCC 27853 and 4 clinical isolates) and 5 *K. pneumoniae* clinical isolates.

Antibiotic sources: Plazomicin was supplied by Achaogen. Amikacin, gentamicin, tobramycin, vancomycin, and levofloxacin were purchased from Sigma. Cefepime and daptomycin were acquired from Bristol Myers Squibb and ProClinical Inc., respectively.

Antimicrobial Susceptibility Testing with Pulmonary Surfactant: Minimum inhibitory concentrations (MICs) were determined using the broth microdilution method recommended by the Clinical and Laboratory Standards Institute (CLSI). The antibiotics tested against the Gram-negative isolates were plazomicin, amikacin, gentamicin, tobramycin, cefepime, and levofloxacin. Additionally, daptomycin and vancomycin were tested against the Gram-positive isolates. Prior to the addition of the antibiotic stock and bacterial inoculate, beractant (Survanta®, Abbott Laboratories), a bovine lung fluid extract, was added to cation adjusted Mueller-Hinton broth (CAMHB) to achieve surfactant concentrations of 1%, 5%, and 10%. Beractant's composition: 25 mg/mL phospholipids, 0.5 -1.75 mg/mL triglycerides, 1.4 - 3.5 mg/mL free fatty acids, and less than 1.0 mg/mL protein (SP-B and SP-C).

Quality control (QC) of antibiotic stocks was established using CLSI QC ranges and CLSI recommended ATCC QC strains, *S. aureus* ATCC 29213 and *E. coli* ATCC 25922.

Changes in antimicrobial activity in the presence of surfactant were expressed in MIC fold change and were calculated by dividing the surfactant MIC by the control (non-surfactant) MIC of the same isolate.

RESULTS

Table 1: MICs of Plazomicin and Comparator Antibiotics vs. *S. aureus* Strains in the Presence of Varying Amounts of Pulmonary Surfactant

Organism	Code	% Surfactant	MIC (µg/mL) Plazomicin	Daptomycin	Amikacin	Gentamicin	Tobramcyin	Vancomycin	Levofloxacin	Cefepime
S. aureus	ATCC 29213	0	0.5	0.25	2	0.25	0.25	1	0.125	2
		1	0.5	8	4	1	0.25	1	0.06	2
		5	1	>16	1	0.5	0.25	1	0.06	2
		10	0.5	>16	4	0.25	0.25	1	0.06	2
	ASAU1014	0	0.25	0.5	0.5	0.125	0.125	2	0.125	>16
		1	0.25	16	0.5	0.125	0.125	2	0.125	>16
		5	0.25	>16	0.5	0.125	0.125	2	0.125	>16
		10	0.125	>16	1	0.125	<=0.06	2	0.125	>16
	ASAU1018	0	1	0.25	2	0.25	0.25	1	0.06	4
		1	0.5	8	2	0.25	0.25	1	0.06	2
		5	0.5	>16	1	0.25	0.25	2	0.06	2
		10	0.25	>16	1	0.125	0.125	1	0.06	8
	ASAU1020	0	1	0.25	2	0.25	0.25	1	0.125	2
		1	0.5	8	1	0.25	0.25	1	0.125	2
		5	0.5	>16	2	0.25	0.25	1	0.125	2
		10	0.5	>16	1	0.25	0.25	1	0.06	2
	ASAU1025	0	1	0.25	2	0.5	0.25	1	0.06	2
		1	0.5	8	2	0.5	0.25	1	0.06	2
		5	0.5	>16	4	0.5	0.25	1	0.06	2
		10	0.5	>16	2	0.25	0.25	1	0.06	2
	ASAU1034	0	0.5	0.25	1	0.25	0.25	1	0.06	>16
		1	0.5	8	2	0.5	0.25	1	0.06	16
		5	2	>16	1	0.125	0.25	0.5	0.06	>16
		10	0.5	>16	2	0.25	0.25	1	0.06	>16
	ASAU1051	0	1	0.5	4	1	1	1	0.06	16
		1	1	16	2	1	2	1	0.06	8
		5	2	>16	2	2	1	1	0.06	8
		10	1	>16	2	0.5	0.5	1	0.06	>16
	ASAU1058	0	1	0.5	2	1	0.25	1	0.125	2
		1	1	8	2	0.5	0.25	1	0.125	2
		5	0.5	>16	2	0.5	0.25	1	0.125	1
		10	0.5	>16	2	0.5	0.25	1	0.25	1
	ASAU1065	0	1	0.25	2	1	0.25	1	0.25	>16
		1	1	8	2	0.25	0.25	1	0.25	>16
		5	0.5	>16	1	0.25	0.25	1	0.25	>16
		10	0.5	>16	1	1	0.25	1	0.5	>16
	ASAU1066	0	0.25	0.5	1	0.125	0.125	2	2	>16
		1	0.25	16	2	0.125	0.25	2	2	>16
		5	0.25	>16	2	0.25	0.25	2	2	>16
		10	0.25	>16	2	0.125	0.25	2	2	>16

Table 2: MICs of Plazomicin and Comparator Antibiotics vs. Gram-Negative Strains in the Presence of Varying Amounts of Pulmonary Surfactant

Organism	Code	% Surfactant	MIC (µg/mL) Plazomicin	Amikacin	Gentamicin	Tobramcyin	Levofloxacin	Cefepime
P. aeruginosa	ATCC 27853	0	2	2	0.5	0.25	2	2
		1	2	2	0.5	0.25	2	1
		5	2	2	0.5	0.25	2	2
		10	2	2	0.5	0.25	2	1
	APAE1004	0	2	2	1	0.25	0.5	2
		1	2	2	1	0.25	0.5	2
		5	2	2	1	0.5	0.5	2
		10	2	2	1	0.25	0.5	2
	APAE1019	0	2	2	1	0.5	0.5	2
		1	2	2	1	0.25	0.5	2
		5	4	4	1	0.25	1	1
		10	2	2	2	0.5	1	1
	APAE1021	0	4	2	0.5	0.5	1	1
		1	4	4	1	0.25	1	1
		5	2	4	1	0.25	1	1
		10	2	4	0.5	0.25	1	1
	APAE1026	0	2	2	1	0.25	4	4
		1	2	2	1	0.25	4	4
		5	2	2	1	0.25	4	4
		10	2	2	1	0.25	4	4
K. pneumoniae	AKPN1021	0	1	2	0.5	0.5	0.06	0.06
		1	0.5	2	0.5	0.5	0.06	0.06
		5	0.5	2	0.5	0.25	0.06	0.03
		10	1	2	0.5	0.5	0.06	0.06
	AKPN1031	0	0.25	1	0.25	0.25	0.5	2
		1	0.25	2	0.25	0.25	1	1
		5	0.25	1	0.25	0.25	1	2
		10	0.25	2	0.25	0.25	0.5	4
	AKPN1034	0	0.125	0.5	0.125	0.125	8	0.06
		1	0.125	2	0.25	0.125	8	0.06
		5	0.125	0.5	0.125	0.125	8	0.06
		10	0.125	1	0.125	0.125	8	0.06
	AKPN1040	0	0.25	2	0.25	0.25	>16	0.125
		1	0.25	2	0.25	0.25	>16	0.06
		5	0.25	2	0.5	0.25	>16	0.06
		10	0.25	1	0.25	0.25	>16	0.125
	AKPN1046	0	0.25	4	0.25	0.25	0.125	0.06
		1	0.5	1	0.5	0.25	0.125	0.06
		5	0.5	2	0.5	0.25	0.25	0.06
		10	0.25	1	0.5	0.25	0.125	0.03

Figure 1: MIC Distribution of Plazomicin and Daptomycin with Increasing Concentrations of Pulmonary Surfactant

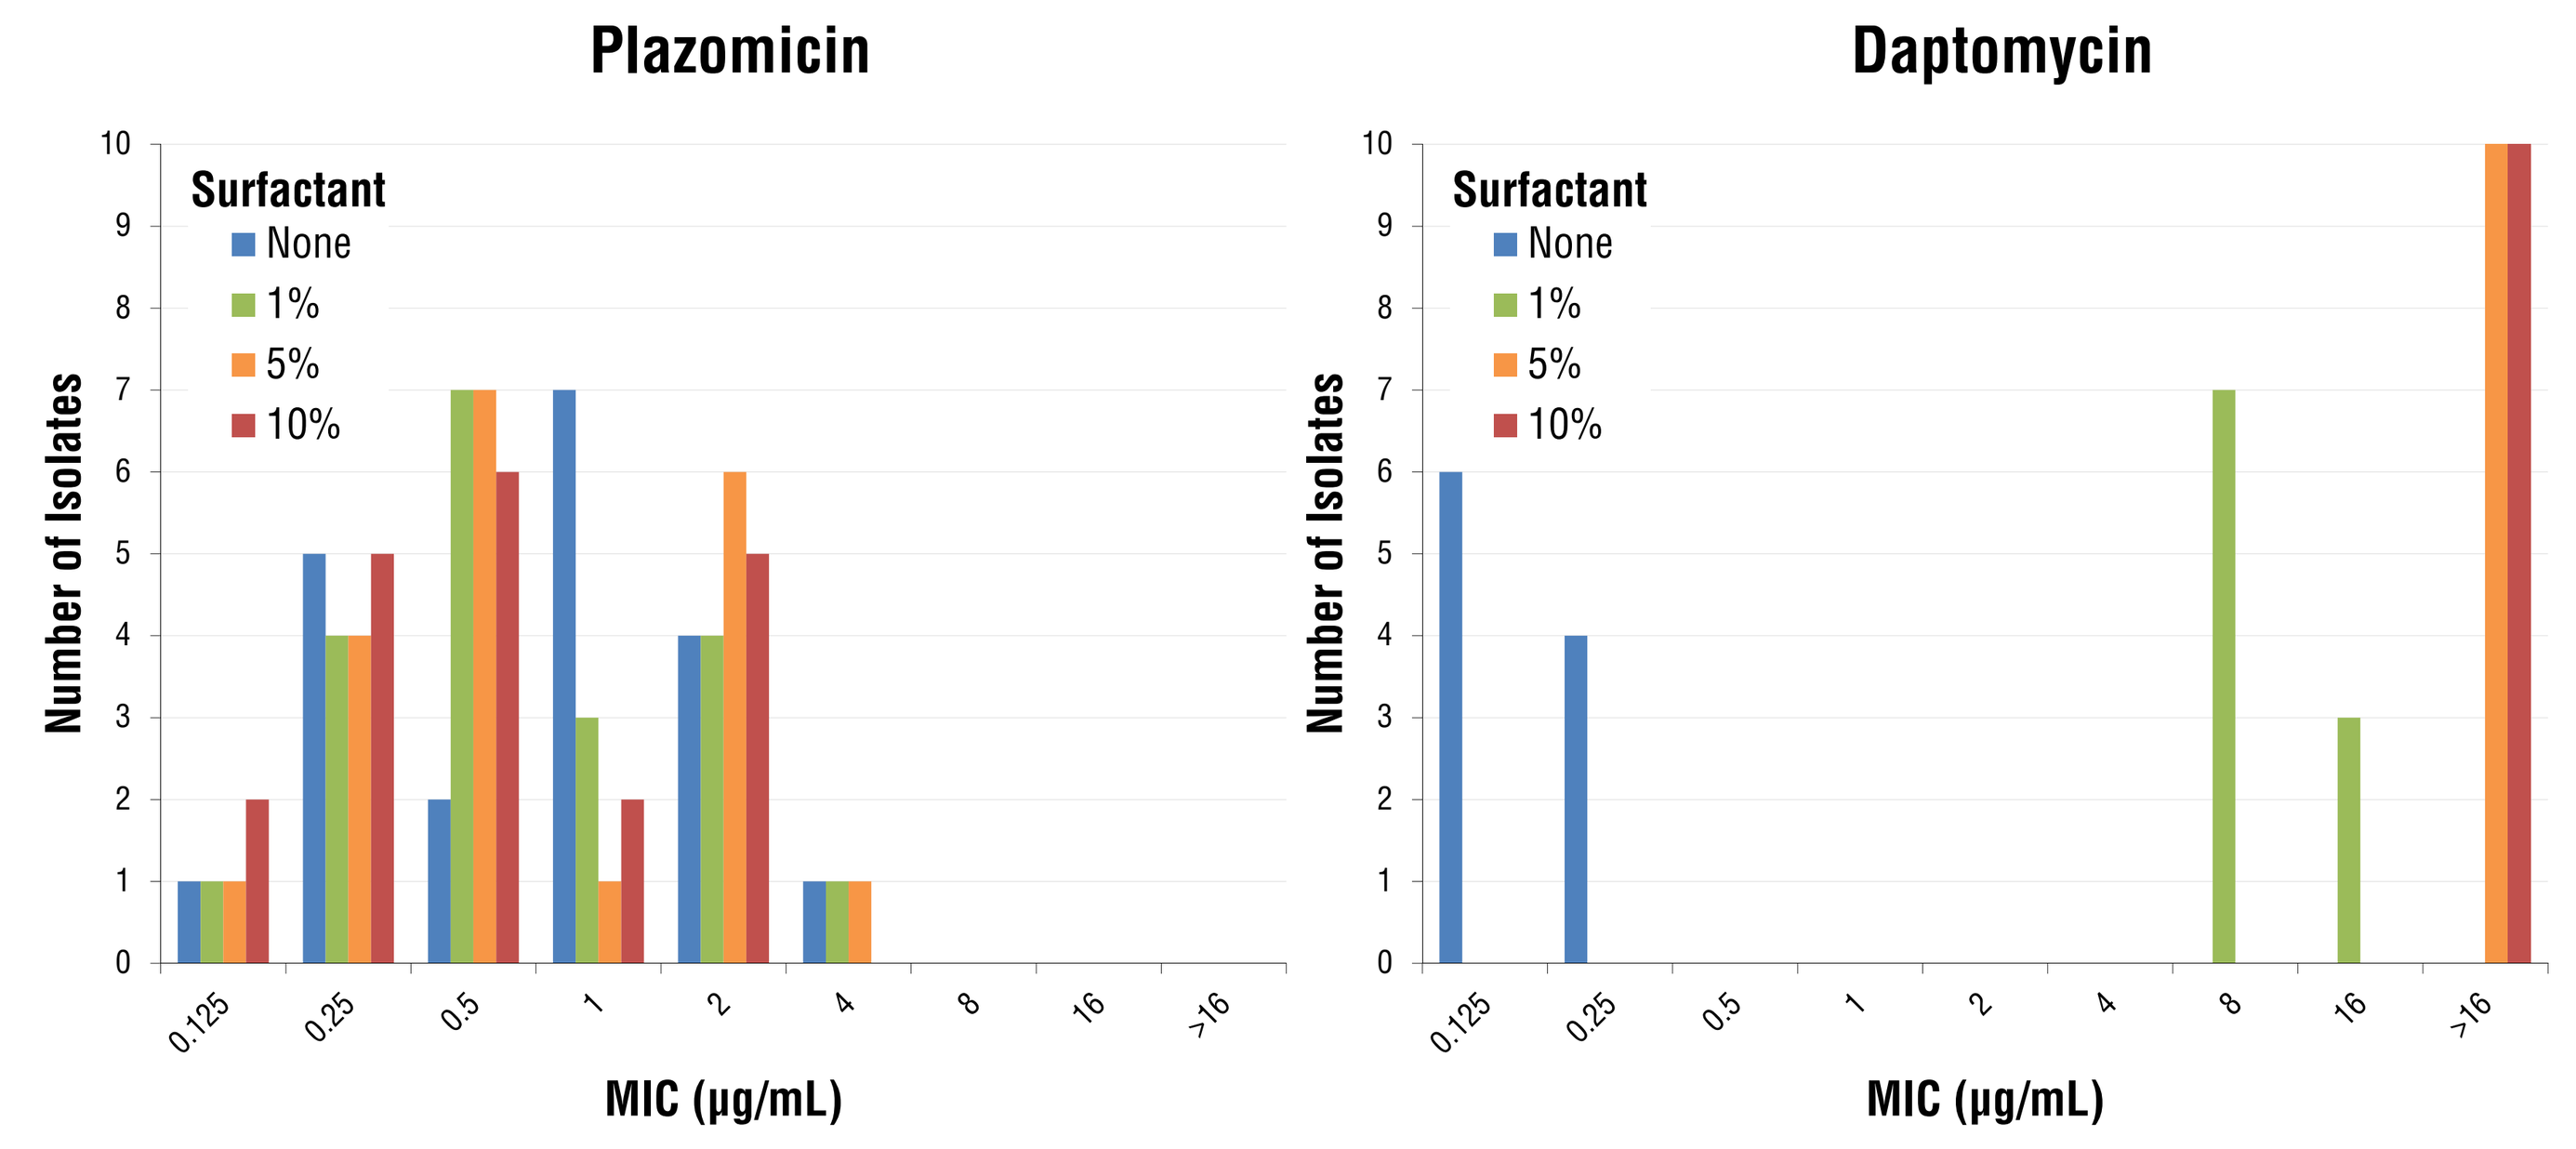

Plazomicin
Daptomycin
Surfactant
None
1%
5%
10%
Number of Isolates
0
1
2
3
4
5
6
7
8
9
10
0.125
0.25
0.5
1
2
4
8
16
>16
MIC (µg/mL)

Table 3: Fold Change in the MIC of Plazomicin and Comparator Antibiotics vs. *S. aureus* Strains in the Presence of Varying Amounts of Pulmonary Surfactant Relative to the MIC in the Absence of Pulmonary Surfactant (MICsurfactant/MICno surfactant)

Organism	Code	% Surfactant	MIC Fold Change Plazomicin	Daptomycin	Amikacin	Gentamicin	Tobramcyin	Vancomycin	Levofloxacin	Cefepime
S. aureus	ATCC 29213	1	1	32	2	4	1	1	0.5	1
		5	2	>64	0.5	2	1	1	0.5	1
		10	1	>64	2	1	1	1	0.5	1
	ASAU1014	1	1	32	1	1	1	1	1	N/A
		5	1	>32	1	1	1	1	1	N/A
		10	0.5	>32	2	1	<=0.5	1	1	N/A
	ASAU1018	1	0.5	32	1	1	1	1	1	0.5
		5	0.5	>64	0.5	1	1	2	1	0.5
		10	0.25	>64	0.5	0.5	0.5	1	1	2
	ASAU1020	1	0.5	32	0.5	1	1	1	1	1
		5	0.5	>64	1	1	1	1	1	1
		10	0.5	>64	0.5	1	1	1	0.5	1
	ASAU1025	1	0.5	32	1	1	1	1	1	1
		5	0.5	>64	2	1	1	1	1	1
		10	0.5	>64	1	0.5	1	1	1	1
	ASAU1034	1	1	32	2	2	1	1	1	<=0.5
		5	4	>64	1	0.5	1	0.5	1	N/A
		10	1	>64	2	1	1	1	1	N/A
	ASAU1051	1	1	32	0.5	1	2	1	1	0.5
		5	2	>32	0.5	2	1	1	1	0.5
		10	1	>32	0.5	0.5	0.5	1	1	>=2
	ASAU1058	1	1	16	1	0.5	1	1	1	1
		5	0.5	>32	1	0.5	1	1	1	0.5
		10	0.5	>32	1	0.5	1	1	2	0.5
	ASAU1065	1	1	32	1	0.25	1	1	1	N/A
		5	0.5	>64	0.5	0.25	1	1	1	N/A
		10	0.5	>64	0.5	1	1	1	2	N/A
	ASAU1066	1	1	32	2	1	2	1	1	N/A
		5	1	>32	2	2	2	1	1	N/A
		10	1	>32	2	1	2	1	1	N/A
MIC Fold Change Range			**0.25 - 4**	**16 - >64**	**0.5 - 2**	**0.25 - 4**	**<=0.5 - 2**	**0.5 - 2**	**0.5 - 2**	**<=0.5 - >=2**

N/A = Not applicable

Table 4: Fold Change in the MIC of Plazomicin and Comparator Antibiotics vs. Gram-Negative Strains in the Presence of Varying Amounts of Pulmonary Surfactant Relative to the MIC in the Absence of Pulmonary Surfactant (MICsurfactant/MICno surfactant)

Organism	Code	% Surfactant	MIC Fold Change Plazomicin	Amikacin	Gentamicin	Tobramcyin	Levofloxacin	Cefepime
P. aeruginosa	ATCC 27853	1	1	1	1	1	1	0.5
		5	1	1	1	1	1	1
		10	1	1	1	1	1	0.5
	APAE1004	1	1	1	1	1	1	1
		5	1	1	1	2	1	1
		10	1	1	1	1	1	1
	APAE1019	1	1	1	1	0.5	1	1
		5	2	2	1	0.5	2	0.5
		10	1	1	2	1	2	0.5
	APAE1021	1	1	2	2	0.5	1	1
		5	0.5	2	2	0.5	1	1
		10	0.5	2	1	0.5	1	1
	APAE1026	1	1	1	1	1	1	1
		5	1	1	1	1	1	1
		10	1	1	1	1	1	1
K. pneumoniae	AKPN1021	1	0.5	1	1	1	1	1
		5	0.5	1	1	0.5	1	0.5
		10	1	1	1	1	1	1
	AKPN1031	1	1	2	1	1	2	0.5
		5	1	1	1	1	2	1
		10	1	2	1	1	1	2
	AKPN1034	1	1	4	2	1	1	1
		5	1	1	1	1	1	1
		10	1	2	1	1	1	1
	AKPN1040	1	1	1	1	1	N/A	0.5
		5	1	1	2	1	N/A	0.5
		10	1	0.5	1	1	N/A	1
	AKPN1046	1	2	0.25	2	1	1	1
		5	2	0.5	2	1	2	1
		10	1	0.25	2	1	1	0.5
MIC Fold Change Range			**0.5 - 2**	**0.25 - 4**	**1 - 2**	**0.5 - 2**	**1 - 2**	**0.5 - 2**

N/A = Not applicable

CONCLUSIONS

- Using an *in vitro* model with bovine-derived lung extract to mimic the lung environment, there was no pulmonary surfactant-related decrease in plazomicin activity when tested at concentrations up to 10% against Gram-negative and Gram-positive organisms.
 - Daptomycin, previously shown to be inactivated by this pulmonary surfactant, showed a marked decrease in activity in the presence of surfactant, demonstrated by MIC fold shifts from 16- to 64-fold in the presence of 1% surfactant, the lowest concentration tested.
- None of the 6 comparators (amikacin, gentamicin, tobramycin, cefepime, levofloxacin or vancomycin) tested showed a surfactant-related decrease in potency.
 - The range of MIC fold change for plazomicin was from 0.25- to 4-fold and was the same as the range for the 3 comparator aminoglycosides in the 20 strain panel.
- The persistence of the antimicrobial activity of plazomicin in the presence of pulmonary surfactant along with its broad spectrum activity warrants consideration for clinical development for the treatment of serious respiratory tract infections.

REFERENCES

1. **Silverman JA, Mortin LI, VanPraagh DG, et al.** Inhibition of daptomycin by pulmonary surfactant: in vitro modeling and clinical impact. J Infect Dis 2005;191:2149-52.

2. **Aggen JB, Armstrong ES, Goldblum AA, et al.** Synthesis and spectrum of the neoglycoside ACHN-490. Antimicrob Agents Chemother 2010;54:4636-42.

3. **Landman D, Babu E, Shah N, et al.** Activity of a novel aminoglycoside, ACHN-490, against cinical isolates of *Escherichia coli* and *Klebsiella pneumoniae* from New York City. J Antimicrob Chemother 2010;65:2123.

4. **Landman D, Kelly P, Bäcker M, et al.** Antimicrobial activity of a novel aminoglycoside, ACHN-490, against *Acinetobacter baumannii* and *Pseudomonas aeruginosa* from New York City. J Antimicrob Chemother 2011;66:332-4.